Safety and Tolerability of Magrolimab Combinations in Patients With Relapsed/Refractory Multiple Myeloma: Safety Run-in Results From a Phase 2 Study

3383

GS-US-558-5915

Barry Paul,[1] Jiri Minarik,[2] Francesca Cottini,[3] Cristina Gasparetto,[4] Jack Khouri,[5] Mitul Gandhi,[6] Jens Hillengass,[7] Moshe Levy,[8] Michaela Liedtke,[9] Sudhir Manda,[10] Irwindeep Sandhu,[11] Douglas Sborov,[12] Ivan Spicka,[13] Saad Usmani,[14] Lin Gu,[15] Michelle Robeson,[15] Michael Murphy,[16] Camille Renard,[15] Christine Chen,[17] Ludek Pour[18]

[1]Levine Cancer Institute, Charlotte, NC, USA; [2]Fakultní Nemocnice Olomouc, Olomouc, Czech Republic; [3]The Ohio State University, Columbus, OH, USA; [4]Duke Cancer Institute, Durham, NC, USA; [5]Cleveland Clinic, Cleveland, OH, USA; [6]US Oncology Site, Virginia Cancer Specialists, Fairfax, VA, USA; [7]Roswell Park Cancer Institute, Buffalo, NY, USA; [8]Baylor University Medical Center, Dallas, TX, USA; [9]Stanford Cancer Center, Palo Alto, CA, USA; [10]Arizona Oncology Associates, Tucson, AZ, USA; [11]Cross Cancer Institute, Edmonton, AB, Canada; [12]Huntsman Cancer Institute at the University of Utah, Salt Lake City, UT, USA; [13]Charles University Hospital Kralovske Vinohrady, Prague, Czech Republic; [14]Memorial Sloan-Kettering Cancer Center, New York, NY, USA; [15]Gilead Sciences, Inc, Foster City, CA, USA; [16]NHS Blood and Transplant and University of Oxford, Oxford, UK; [17]Princess Margaret Cancer Centre, Toronto, ON, Canada; [18]University Hospital Brno, Brno, Czech Republic

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- In heavily pretreated patients with multiple myeloma who relapsed or were refractory to more than 3 prior lines of therapy, magrolimab demonstrated an acceptable safety profile and showed minimal additive toxicity when given in combination with daratumumab, pomalidomide/dexamethasone, or carfilzomib/ dexamethasone
- Transient on-target anemia was observed after the first dose of magrolimab but was rarely observed with subsequent doses
- Criteria for a recommended phase 2 dose (RP2D) were met for the dose expansion portion of the study in each combination

Plain Language Summary

Patients with multiple myeloma (MM) that has returned or is resistant to the last treatment tend to get sicker and die sooner, demonstrating a need for better treatments. Researchers have found that combinations of drugs that work in different ways may be better for the treatment of MM. We are reporting on the safety of 3 different drug combinations, each containing a new drug called magrolimab. Myeloma cells have a number of “don’t eat me” signals that enable them to escape death from the patient's immune system. Magrolimab blocks these signals. The other drugs combined with magrolimab are already commonly used to treat MM that is resistant to prior treatment or has returned. We found that all 3 magrolimab-drug combinations were relatively safe and that adding magrolimab does not make the combination treatment substantially more toxic.

References: **1.** Chari A, et al. *N Engl J Med*. 2019;381(8):727–738. **2.** Bruno AS, et al. *Expert Rev Hematol*. 2020;13(9):1017–1025. **3.** Drug therapy for multiple myeloma. American Cancer Society. Revised August 15, 2023. Accessed October 17, 2023. http://www.cancer.org/cancer/multiple-myeloma/treating/chemotherapy.html. **4.** Liu J, et al. *PLOS ONE*. 2015;10(9):e0137345. **5.** Chao MP, et al. *Front Oncol*. 2020;9:1380. **6.** Kim D, et al. *Leukemia*. 2012;26(12):2538–2545. **7.** Sun J, et al. *Cancers (Basel)*. 2020;12(2):305. **8.** Gilead Sciences, Inc. Data on file.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Editing and production assistance were provided by Ashfield MedComms, an Inizio Company, and funded by Gilead Sciences, Inc.

Disclosures: Disclosures can be found by scanning the QR code in the upper corner of the poster.

Correspondence: Barry Paul, barry.paul@atriumhealth.org

Introduction

- Treatment options remain limited for heavily pretreated patients with relapsed/refractory multiple myeloma (RRMM) who have already received the main classes of therapy (including proteasome inhibitors and anti–cluster of differentiation 38 [CD38] antibodies)[1,2]
- Novel combination therapies with new drugs possessing unique mechanisms of action and nonoverlapping toxicity are needed to improve patient outcomes[3]
- Magrolimab is a first-in-class, immuno-oncology therapy that blocks CD47, an antiphagocytic signal overexpressed in cancer cells (including MM) that enables them to evade immune surveillance[4,5]
- In vitro blockage of CD47 resulted in increased elimination of MM cells,[6,7] and preclinical data suggest that magrolimab may synergize with commonly used standard-of-care backbone agents in MM[8]
- Reported here are initial safety and tolerability data from 3 safety run-in (SRI) cohorts of our phase 2, open-label, multi-arm study (NCT04892446) in which magrolimab-based combinations were evaluated in patients with RRMM

Methods

- The objective of the SRI portion of the study was to evaluate the safety and tolerability profile and determine the RP2D of the following magrolimab combinations in RRMM:
 - — Magrolimab + daratumumab (Magro + Dara)
 - — Magrolimab + pomalidomide/dexamethasone (Magro + Pd)
 - — Magrolimab + carfilzomib/dexamethasone (Magro + Kd)
- The study designs for each SRI cohort are illustrated in **Figure 1**

Figure 1. Study Design

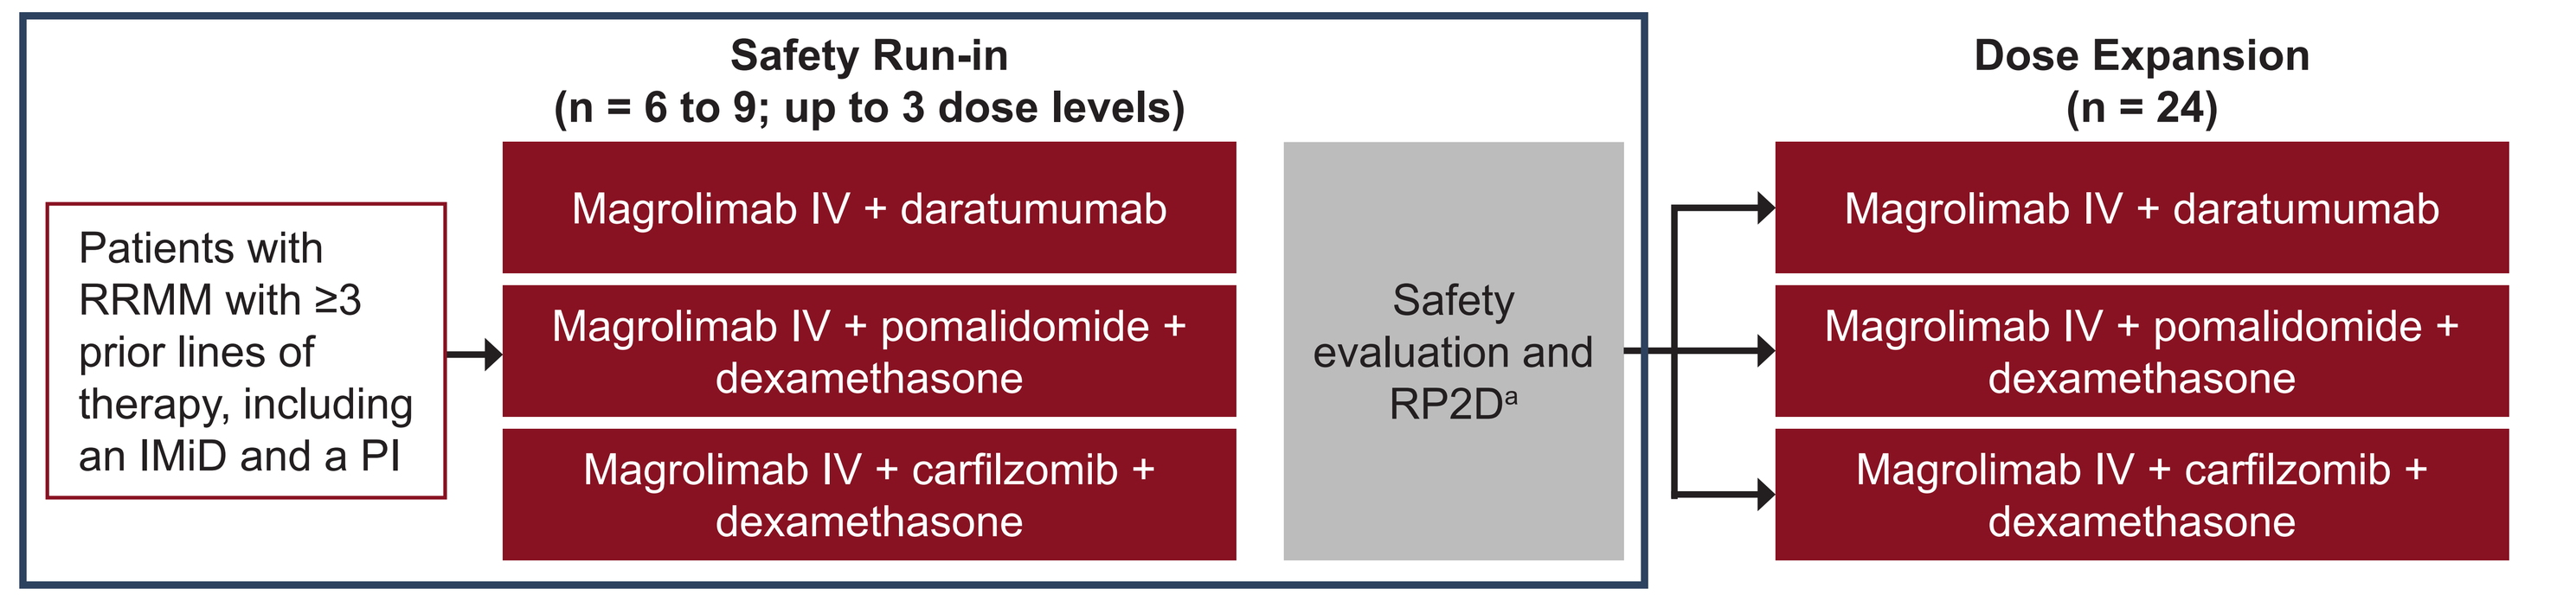

Safety Run-in
(n = 6 to 9; up to 3 dose levels)
Patients with RRMM with ≥3 prior lines of therapy, including an IMiD and a PI
Magrolimab IV + daratumumab
Magrolimab IV + pomalidomide + dexamethasone
Magrolimab IV + carfilzomib + dexamethasone
Safety evaluation and RP2D[a]
Dose Expansion
(n = 24)
Magrolimab IV + daratumumab
Magrolimab IV + pomalidomide + dexamethasone
Magrolimab IV + carfilzomib + dexamethasone

[a]To select an RP2D, ≤2 of 6 DLT-evaluable patients could experience a DLT. DLTs were evaluated throughout cycle 1 (35 days) and defined as any investigator-assessed grade ≥3 TEAEs that worsened from baseline during the DLT assessment period and were at least possibly related to magrolimab. Exceptions to this definition included grade 3 anemia or grade 3/4 lymphopenia or leukopenia that was not medically significant; grade 3 neutropenia that resolved to grade ≤2 within 2 weeks; grade 3 hyperbilirubinemia, fatigue, hypermagnesemia, isolated electrolyte abnormalities, and elevation in alanine aminotransferase/aspartate aminotransferase/alkaline phosphatase that resolved to grade ≤2 within 1 week; grade 3 tumor lysis syndrome, electrolyte disturbances (hyperkalemia, hypophosphatemia, hyperuricemia), nausea, vomiting, or diarrhea that resolved to grade ≤2 within 72 hours; and grade 3 magrolimab infusion reactions in the absence of an optimal pretreatment regimen.
DLT, dose-limiting toxicity; IMiD, immunomodulatory drug; IV, intravenous; PI, proteasome inhibitor; TEAE, treatment-emergent adverse event.

- At the initial dose level tested, magrolimab was given intravenously as a 1-mg/kg priming dose on day 1 of cycle 1, followed by a maintenance dose of 30 mg/kg once every week during cycles 1 and 2, then once every 2 weeks in cycles 3+; all other agents were administered at standard doses and schedules per clinical guidelines (**Table 1**)

Table 1. Dosing and Treatment Schedule for the SRI Cohorts

Drug	Dose	Cycle 1 (35 days)	Cycle 2 (28 days)	Cycle 3+ (28 days/cycle)
Magrolimab[a]	Priming dose: 1 mg/kg IV	Week 1, day 1	–	–
	Starting dose: 30 mg/kg IV	Days 8, 15, 22, 29	Every week on days 1, 8, 15, 22	Days 1 and 15
Daratumumab[a]	16 mg/kg IV or 1800 mg SC	Days 8, 15, 22, 29	Every week on days 1, 8, 15, 22	Days 1, 15 (every 2 weeks) for cycles 3–6 (8 doses total), followed by day 1 (every 4 weeks) of cycles 7+
Pomalidomide	4 mg PO	Days 1–21 (daily)	Days 1–21 (daily)	Days 1–21 (daily)
Dexamethasone[b]	40 mg PO	Days 1, 8, 15, 22, 29	Days 1, 8, 15, 22	Days 1, 8, 15, 22
Carfilzomib[c]	20/70 mg/m² IV	Days 8 (20 mg/m²), 15 (70 mg/m²), 22 (70 mg/m²)	Days 1, 8, 15	Days 1, 8, 15
Dexamethasone[b]	40 mg PO	Days 8, 15, 22, 29	Days 1, 8, 15, 22	Days 1, 8, 15, 22 for cycles 3–9, and days 1, 8, 15 for cycles 10+

[a]Premedication with an antipyretic, antihistamine, and corticosteroid (daratumumab only) was administered to mitigate infusion-related reactions (IRRs). For magrolimab, this regimen was required before administration of the first 4 doses and in case of reintroduction with repriming; premedication during subsequent infusions was continued at the investigator's discretion thereafter but was mandatory if a Grade 3 IRR occurred. For daratumumab, this regimen was administered before all doses. [b]Dexamethasone starting dose is 20 mg in patients >75 years old. [c]Recommended starting dose of carfilzomib is 20 mg/m² on cycle 1, day 8. If tolerated, escalate dose to 70 mg/m² on cycle 1, day 15, and thereafter. PO, orally; SC, subcutaneous.

- Primary end points were incidence of TEAEs, DLTs, and lab abnormalities
 - — TEAEs were evaluated according to National Cancer Institute Common Terminology Criteria for Adverse Events (version 5.0) (NCI CTCAE v5.0)
 - — Patients were included in the DLT-evaluable population if they met 1 of 2 criteria:
 - ○ Experienced a DLT after infusion of the first dose of any study drug and throughout cycle 1, or
 - ○ Completed cycle 1 without a DLT and received ≥3 magrolimab infusions and
 - ○ Magro + Dara: ≥2 doses of daratumumab
 - ○ Magro + Pd: ≥10 doses of pomalidomide and ≥2 doses of dexamethasone
 - ○ Magro + Kd: ≥2 doses of carfilzomib and ≥2 doses of dexamethasone
 - — To select a dose level as RP2D, ≤2 of 6 DLT-evaluable patients could experience a DLT at this dose level; otherwise, the magrolimab dose would be de-escalated and a new cohort assessed
 - — Safety was assessed in patients who received ≥1 dose of any study drug

Results

Patient Demographics and Baseline Characteristics

- Of the 25 total patients treated, all were treated at the initial magrolimab dose level
 - — Nine patients were enrolled in the Magro + Dara and Magro + Pd cohorts, respectively; 7 patients were enrolled in the Magro + Kd cohort (**Table 2**)
- The median (range) number of prior lines of therapy received across all cohorts was 4 (2–9)

Table 2. Patient Demographics and Baseline Characteristics of SRI Cohorts

Characteristic	Magro + Dara (N = 9)	Magro + Pd (N = 9)	Magro + Kd (N = 7)
Age (years), median (range)	59 (55–78)	69 (46–76)	64 (57–82)
Male, n (%)	2 (22)	7 (78)	4 (57)
Race, n (%)			
White	7 (78)	8 (89)	6 (86)
Black	1 (11)	1 (11)	1 (14)
Native Hawaiian or Pacific Islander	1 (11)	0	0
Ethnicity, n (%)			
Not Hispanic or Latino	9 (100)	9 (100)	7 (100)
Hispanic or Latino	0	0	0
ECOG performance status, n (%)			
0	2 (22)	0	2 (29)
1	6 (67)	9 (100)	4 (57)
2	1 (11)	0	1 (14)
Prior lines of therapy, median (range)	6 (4–9)	3 (3–6)	7 (2–8)
R-ISS at screening, n (%)			
I	6 (67)	4 (44)	3 (43)
II	2 (22)	3 (33)	3 (43)
III	1 (11)	2 (22)	1 (14)

ECOG, Eastern Cooperative Oncology Group; R-ISS, Revised International Staging System for Myeloma.

DLTs and Safety Summary

- The mean (SD) number of cycles of magrolimab exposure in the Magro + Dara, Magro + Pd, and Magro + Kd cohorts was 2.0 (2.29), 2.9 (1.62), and 2.3 (1.60), respectively
- Two DLTs occurred during the assessment period: grade 3 febrile neutropenia (Magro + Dara) and grade 3 dyspnea (Magro + Pd), experienced in the context of infusion-related reaction
- All patients experienced a TEAE, and all but 2 had a magrolimab-related TEAE
- Two patients (1 each in Magro + Pd and Magro + Kd) had a TEAE that led to magrolimab discontinuation
 - — Only the Magro + Pd patient discontinued magrolimab treatment because of magrolimab-related TEAEs reported on day 15: grade 1 fatigue, grade 3 decreased neutrophil count, and grade 3 decreased white blood cell count
- No deaths due to TEAEs occurred during the study period

Treatment-Emergent Adverse Events

- The most common TEAE observed in each SRI cohort was anemia; other common TEAEs (>2 patients in any SRI cohort) are shown in **Table 3**

Table 3. TEAEs in >2 Patients in Any SRI Cohort

Patients, n/N (%)	Magro + Dara		Magro + Pd		Magro + Kd	
	Any grade	Grade 3-4	Any grade	Grade 3-4	Any grade	Grade 3-4
Anemia	6/9 (66.7)	3/9 (33.3)	5/9 (55.6)	2/9 (22.2)	3/7 (42.9)	1/7 (14.3)
Fatigue	4/9 (44.4)	1/9 (11.1)	3/9 (33.3)	1/9 (11.1)	1/7 (14.3)	0/7 (0)
Headache	4/9 (44.4)	0/9 (0)	0/9 (0)	0/9 (0)	1/7 (14.3)	0/7 (0)
Infusion-related reaction	3/9 (33.3)	0/9 (0)	0/9 (0)	0/9 (0)	1/7 (14.3)	0/7 (0)
Nausea	3/9 (33.3)	0/9 (0)	1/9 (11.1)	0/9 (0)	0/7 (0)	0/7 (0)
Decreased neutrophil count	1/9 (11.1)	0/9 (0)	3/9 (33.3)	3/9 (33.3)	1/7 (14.3)	1/7 (14.3)
Dyspnea	1/9 (11.1)	0/9 (0)	3/9 (33.3)	1/9 (11.1)	0/7 (0)	0/7 (0)

TEAEs graded according to NCI CTCAE (v5.0). Data are presented as n patients exhibiting the event out of the total N patients.

Magrolimab-Related Treatment-Emergent Adverse Events

- The most common magrolimab-related TEAEs (>2 patients in any SRI cohort) were anemia, fatigue, and thrombocytopenia (**Table 4**)
 - — Magrolimab-related anemia was reported in 13 out of 25 patients (grade ≥3, n = 6)
 - — The median (interquartile range [IQR]) drop in hemoglobin level after the magrolimab priming dose (1 mg/kg on cycle 1, day 1) ranged from 0.6 (0.1 to 1.5) to 2.0 (1.3 to 2.3) g/dL across the 3 cohorts; the median (IQR) drops after the second magrolimab dose (30 mg/kg on cycle 1, day 8) were lower and ranged from 0.0 (−0.1 to 0.5) to 0.7 (0.3 to 1.1) g/dL across cohorts (**Table 5**)
- Grade 3-4 magrolimab-related TEAEs reported in >1 patient in any SRI cohort were anemia (Magro + Dara), decreased neutrophil count (Magro + Pd), and decreased platelet count (Magro + Pd)

Table 4. Magrolimab-Related TEAEs in >1 Patient in Any SRI Cohort

Patients, n/N (%)	Magro + Dara		Magro + Pd		Magro + Kd	
	Any grade	Grade 3-4	Any grade	Grade 3-4	Any grade	Grade 3-4
Anemia	6/9 (66.7)	3/9 (33.3)	4/9 (44.4)	2/9 (22.2)	3/7 (42.9)	1/7 (14.3)
Fatigue	3/9 (33.3)	0/9 (0)	3/9 (33.3)	1/9 (11.1)	1/7 (14.3)	0/7 (0)
Thrombocytopenia	1/9 (11.1)	0/9 (0)	2/9 (22.2)	1/9 (11.1)	2/7 (28.6)	1/7 (14.3)
Decreased neutrophil count	1/9 (11.1)	0/9 (0)	2/9 (22.2)	2/9 (22.2)	1/7 (14.3)	1/7 (14.3)
Decreased platelet count	1/9 (11.1)	0/9 (0)	2/9 (22.2)	2/9 (22.2)	1/7 (14.3)	1/7 (14.3)
Headache	3/9 (33.3)	0/9 (0)	0/9 (0)	0/9 (0)	0/7 (0)	0/7 (0)
Hypokalemia	2/9 (22.2)	0/9 (0)	0/9 (0)	0/9 (0)	0/7 (0)	0/7 (0)
Infusion-related reaction	2/9 (22.2)	0/9 (0)	0/9 (0)	0/9 (0)	1/7 (14.3)	0/7 (0)

TEAEs graded according to NCI CTCAE (v5.0). Data are presented as n patients exhibiting the event out of the total N patients.

Table 5. Summary of Hemoglobin Change From Baseline

Hemoglobin (g/dL)	Magro + Dara (N = 9)	Magro + Pd (N = 9)	Magro + Kd (N = 7)
Baseline			
n	9	9	7
Mean (SD)	10.8 (1.14)	10.7 (1.36)	12.2 (2.20)
Median (IQR)	10.8 (10.0–11.4)	10.7 (9.9–11.3)	12.3 (9.9–13.0)
C1D1: post-dose change from baseline			
n	5	7	6
Mean (SD)	−1.9 (0.68)	−0.7 (0.69)	−1.0 (0.58)
Median (IQR)	−2.0 (−2.3 to −1.3)	−0.6 (−1.5 to −0.1)	−1.1 (−1.4 to −0.9)
C1D8: post-dose change from baseline			
n	4	5	5
Mean (SD)	−0.3 (0.77)	−0.9 (0.99)	0.0 (0.75)
Median (IQR)	−0.6 (−0.7 to 0.2)	−0.7 (−1.1 to −0.3)	0.0 (−0.5 to 0.1)

C, cycle; D, day.

- There were 3 patients with magrolimab-related infusion-related reactions (IRRs; all grade 1-2); 1 patient had an IRR related to subcutaneous daratumumab
- Serious magrolimab-related TEAEs occurred in 5 patients
 - — Magro + Dara: febrile neutropenia, a DLT, on day 12 (n = 1); anemia on day 21 (n = 1); and bacteremia on day 135 (n = 1)
 - ○ The patient with anemia experienced 3 anemia serious TEAEs during the study, including 1 prior to the first dose of study drug
 - — Magro + Pd: dyspnea, a DLT, on day 9, followed by febrile neutropenia on day 22 (n = 1; occurred in the same patient)
 - — Magro + Kd: pneumonia on day 75 (n = 1)

65th American Society of Hematology (ASH) Annual Meeting; December 9-12, 2023; San Diego, California, United States